Evolution in Neutrophil-to-lymphocyte ratio (NLR) among advanced Soft Tissue Sarcoma (STS) patients treated with pazopanib within EORTC 62043/62072 trials

Abstract 4511

De Maio E[1], Touati N[1], Litière S[1], Sleijfer S[2], van der Graaf WT[3], Le Cesne A[4], D'Ambrosio L[5], Casali PG[6], Italiano A[7], Desar I[8], Gronchi A[6]

[1]European Organization for Research and Treatment of Cancer, Brussels, Belgium; [2]Erasmus MC - Cancer Institute, Rotterdam, the Netherlands; [3]The Institute of Cancer Research and the Royal Marsden NHS Foundation Trust, London, UK; [4]Institut Gustave Roussy, Villejuif, France; [5]Candiolo Cancer Institute - FPO, IRCCS, Candiolo, Italy; [6]Fondazione IRCCS Istituto Nazionale dei Tumori, Milano, Italy; [7]Institut Bergonié, Bordeaux, France; [8]Radboud University Medical Center, Nijmegen, the Netherlands

BACKGROUND

A high NLR has been shown to be associated with a poor prognosis in several solid tumors including STS and might be helpful for patient stratification and individual risk assessment. The aims of this study were to confirm that higher NLR at baseline is associated with worse prognosis in STS and to evaluate if an early decline of NLR during treatment with pazopanib is associated with a more favorable prognosis.

PATIENT AND METHODS

The single-arm phase II EORTC 62043 and placebo-controlled phase III EORTC 62072 were both investigating the effect of pazopanib in patients with advanced STS. We evaluated NLR at baseline and 50 days later. Multivariate analyses on pazopanib-treated patients investigated the prognostic value on both Progression-Free Survival (PFS) and Overall Survival (OS) of NLR at baseline as well as the predictive value of changes in NLR from baseline to the 50 days landmark. Sensitivity analyses were conducted on the placebo-treated patients.

PFS duration - N/L ratio at baseline

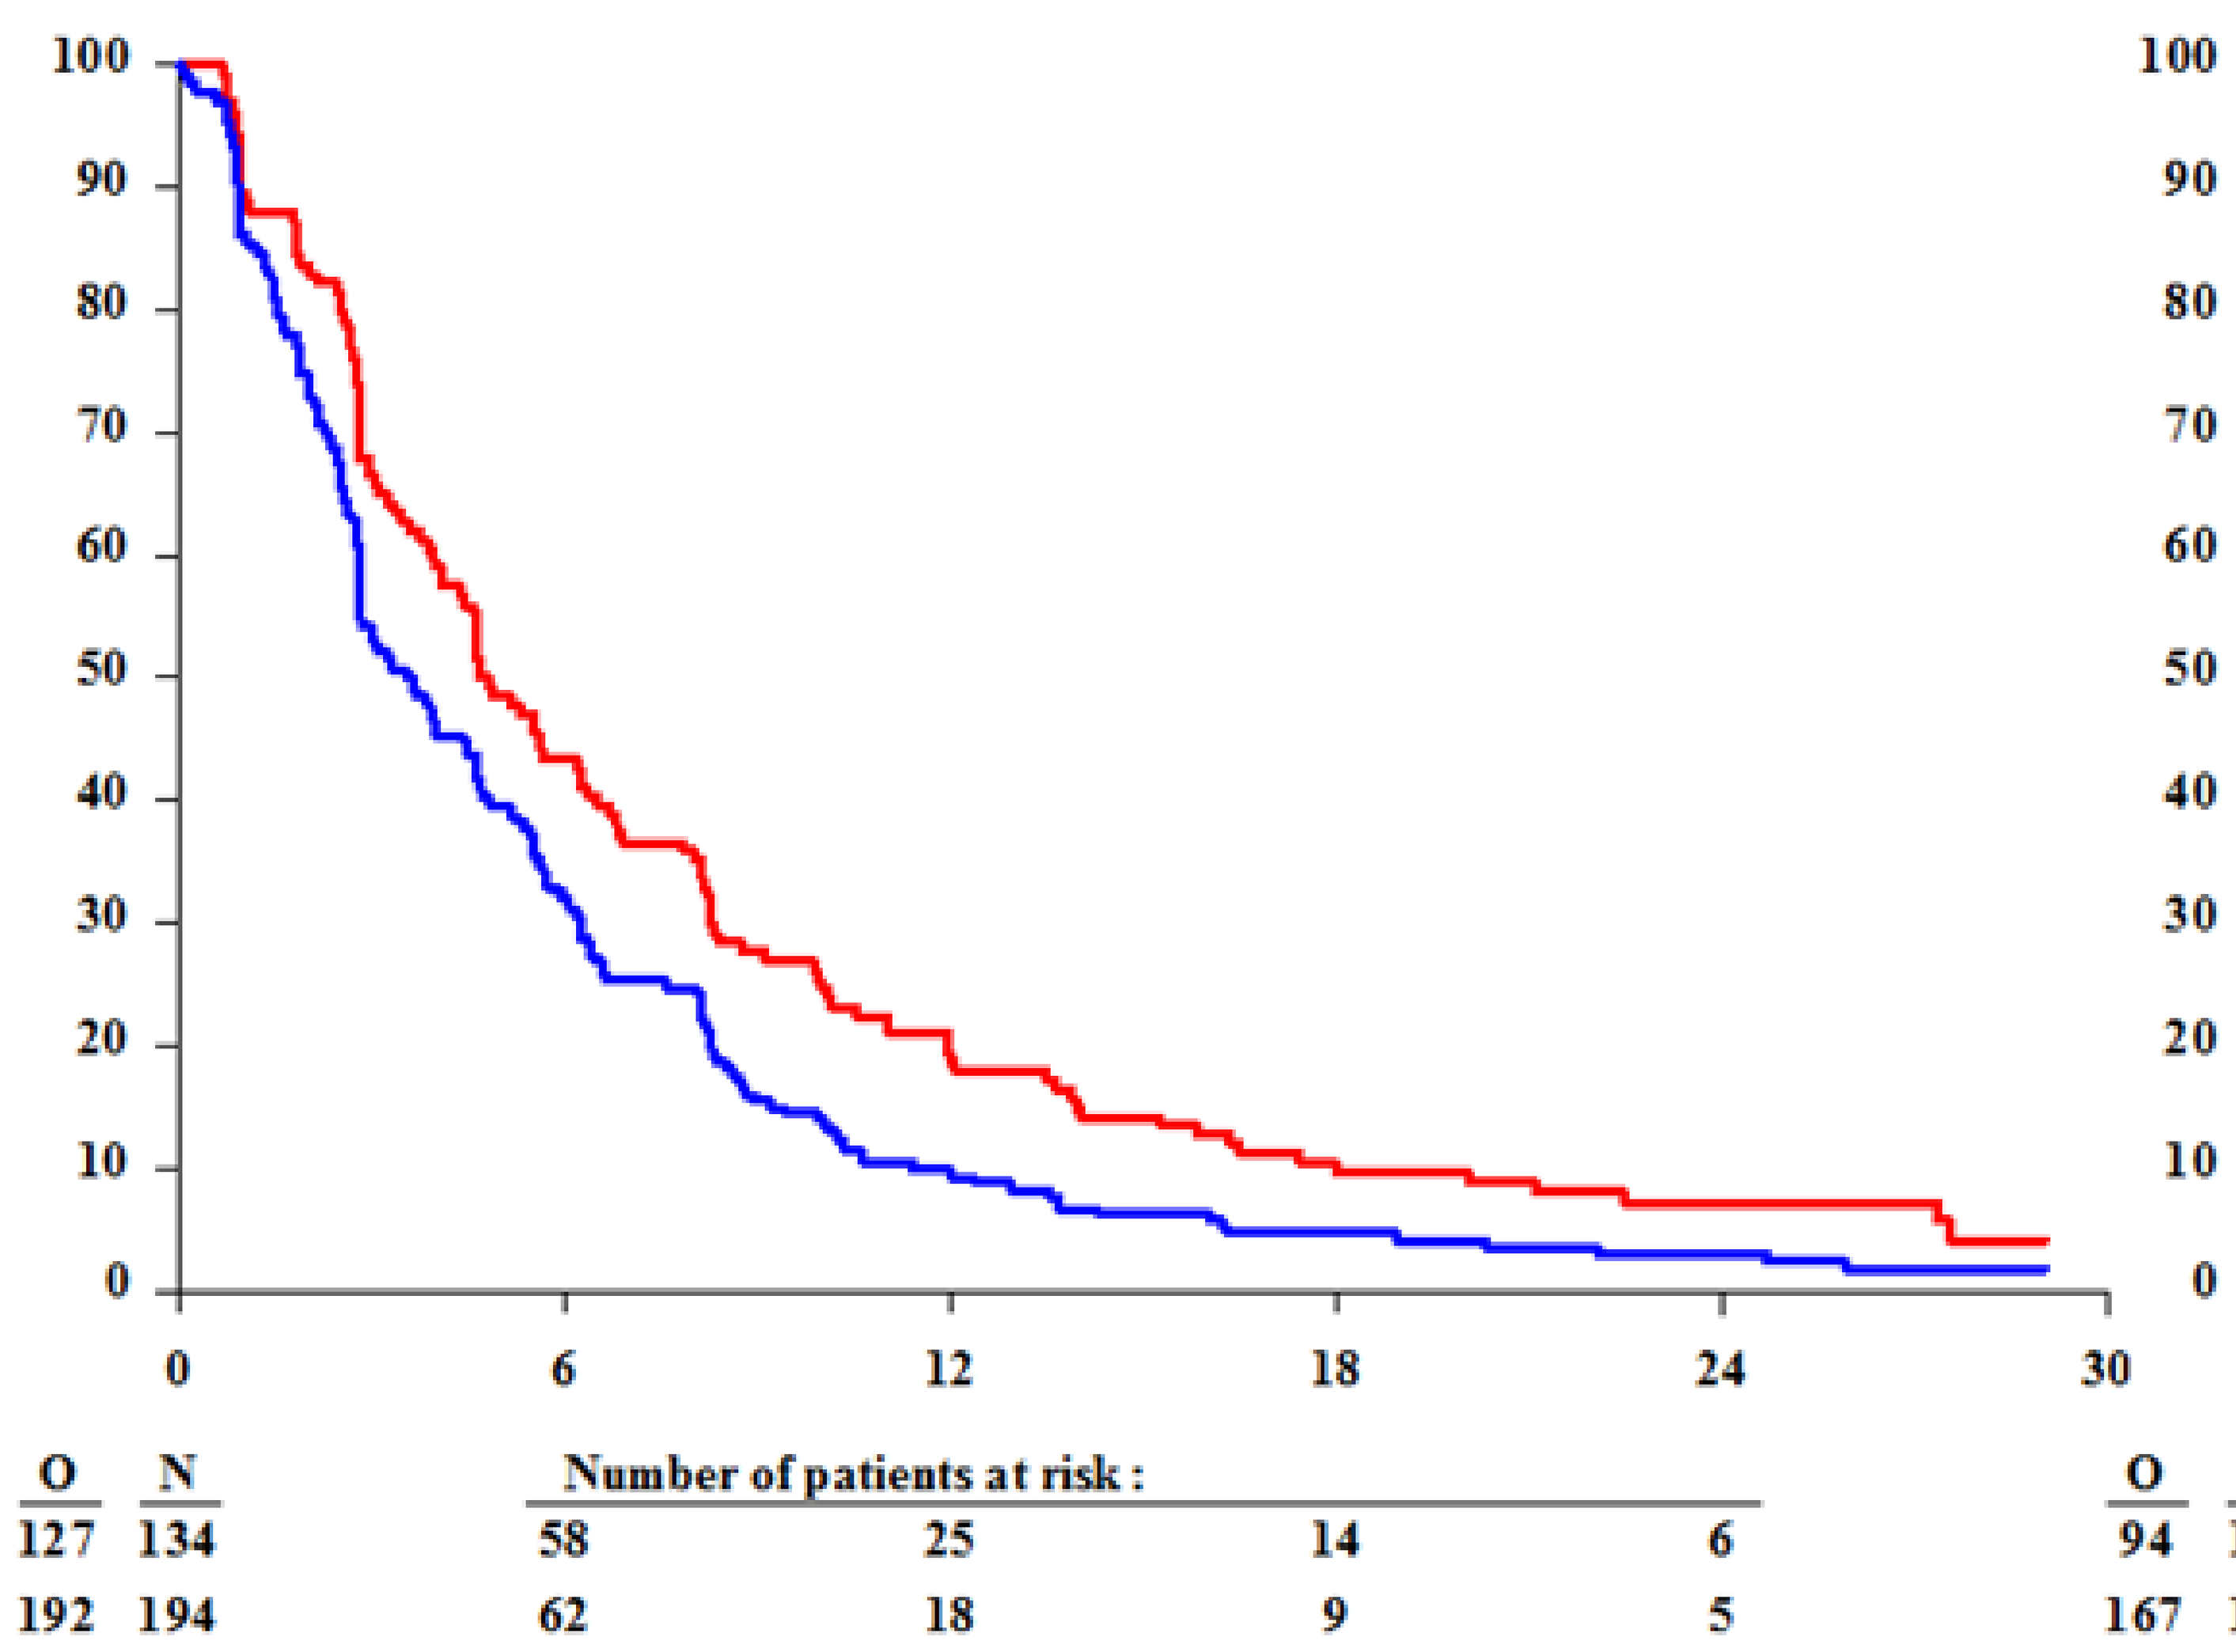
100
90
80
70
60
50
40
30
20
10
0
0
6
12
18
24
30
O N
Number of patients at risk :
127 134 58 25 14 6
192 194 62 18 9 5

OS duration - N/L ratio at baseline

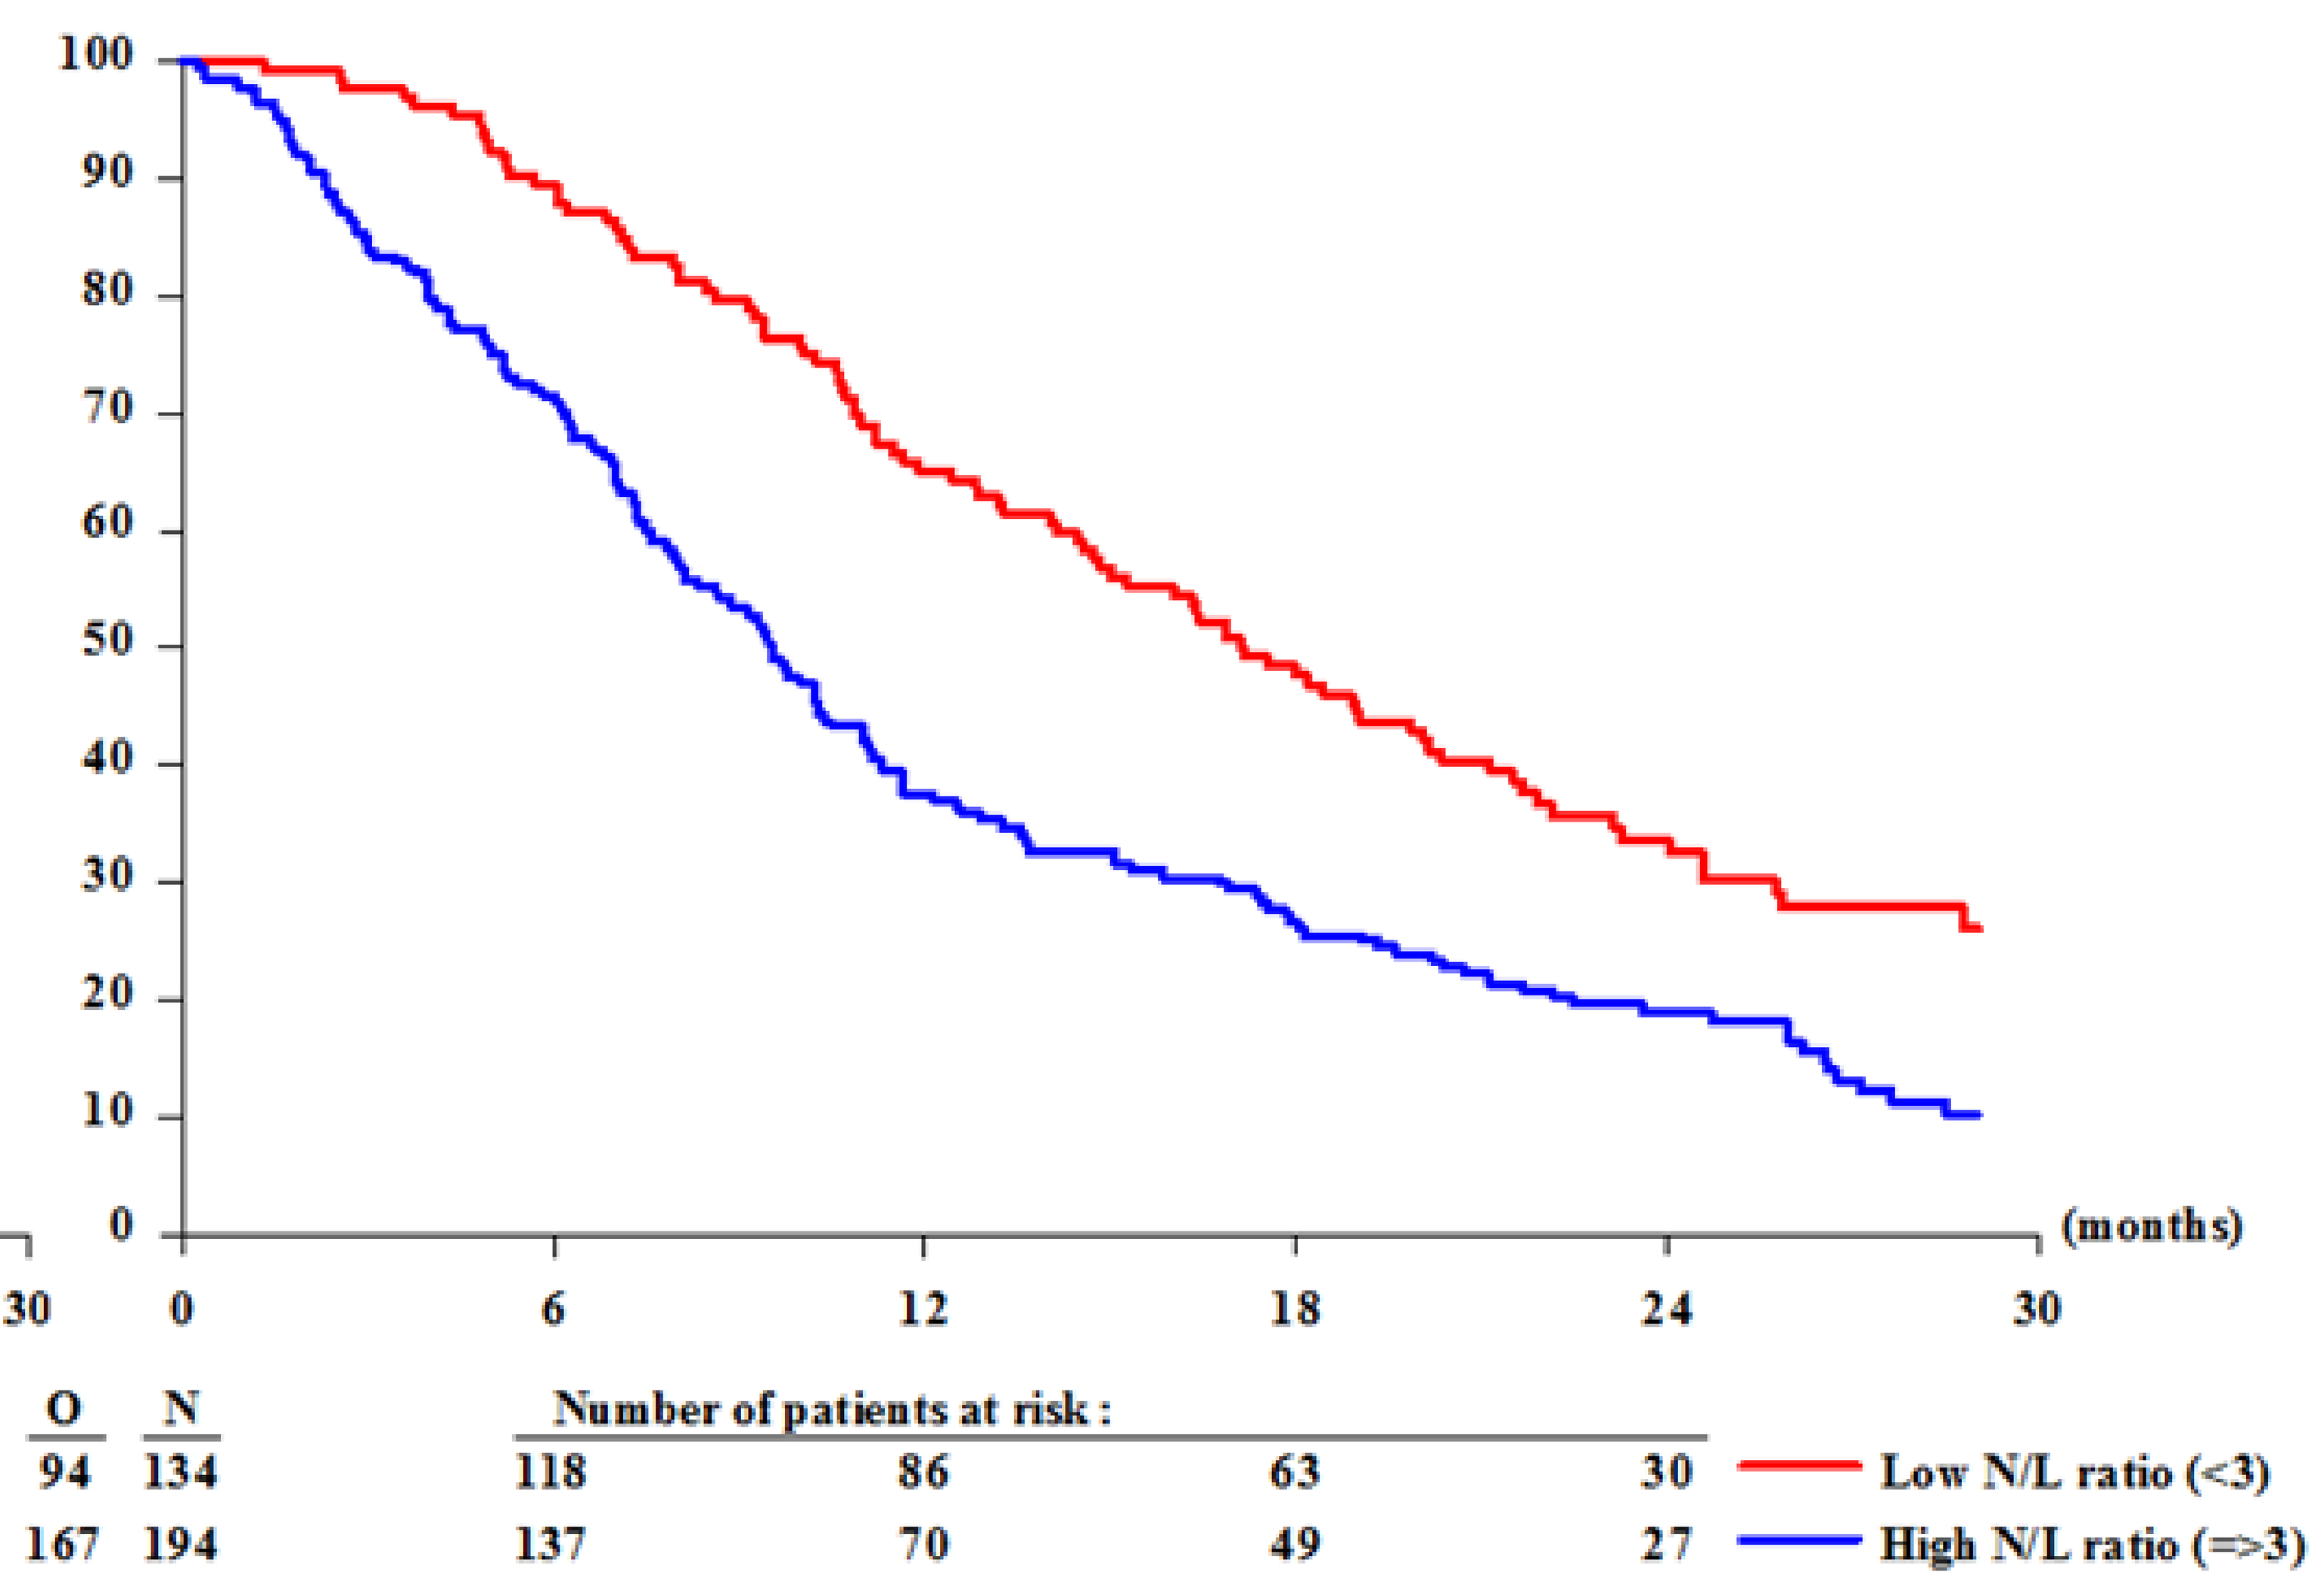
100
90
80
70
60
50
40
30
20
10
0
(months)
0
6
12
18
24
30
O N
Number of patients at risk :
94 134 118 86 63 30
167 194 137 70 49 27
Low N/L ratio (<3)
High N/L ratio (=>3)

Adjusted multivariate Cox models*		Progression-Free Survival: Patients	Observed Events	Median PFS (95% CI, months)	HR (95% CI)	Wald p-value	Overall Survival: Patients	Observed events	Median OS (95% CI, months)	HR (95% CI)	Wald p-value
*Overall population at baseline***		*N=326*	*N=319*				*N=326*	*N=259*			
NLR at baseline	**N/L ratio <3**	134 (41.1)	127 (40.1)	4.6 (4.1, 6.2)	1.00	0.002	134 (41.1)	94 (36.3)	17.0 (14.5, 20.2)	1.00	<0.001
	N/L ratio ≥3	192 (58.9)	190 (59.9)	3.6 (2.8, 4.6)	1.44 (1.14, 1.82)		192 (58.9)	165 (63.7)	9.5 (8.0, 10.9)	1.86 (1.43, 2.41)	
*Population without event at 50 days***		*N=262*	*N=253*				*N=302*	*N=239*			
NLR change	**>40% decrease**	101 (38.5)	97 (38.3)		0.94 (0.71, 1.24)	0.685	108 (35.8)	86 (36.0)		0.96 (0.72, 1.28)	0.255
	No change	143 (54.6)	138 (54.5)		1.00		167 (55.3)	128 (53.6)		1.00	
	>40% increase	18 (6.9)	18 (7.1)		1.19 (0.71, 1.98)		27 (8.9)	25 (10.5)		1.40 (0.90, 2.16)	

**: Adjustment covariates are not displayed in the table (Performance status, Gender, Tumor grade, Histology, Age, history of cardiac event, cardiac AEs, infection AEs)*

***: 2 patients are excluded from multivariate analysis because of missing tumor grade*

RESULTS

Among the 333 eligible patients treated with pazopanib, a NLR at baseline ≥3 - representing 134 patients out of 326 known NLR - was associated with shorter PFS and OS in comparison to NLR <3 (HR 1.44; 95% confidence interval {CI} = 1.14-1.82; p-value 0.002 and HR 1.86; 95% CI = 1.43-2.41; p-value <0.001, respectively). Changes in NLR ratio were defined as a difference of at least 40% with baseline. Compared with no changes, an increase or decrease in NLR did not affect PFS (HR 1.19; 95% CI= 0.71-1.98 and HR 0.94; 95% CI = 0.71-1.24; p-value 0.685, N=262 patients) or OS (HR 1.40; 95% CI = 0.90-2.16 and HR 0.96; 95% CI = 0.7 2- 1.28; p-value 0.255, N=302 patients). Thresholds other than 40% difference to define NLR change did not impact the result and no association between changes in NLR and outcome was seen in placebo-treated patients. The median NLR change in patients treated with pazopanib was a decrease of 30.4% compared to an increase of 14.5% in placebo.

CONCLUSION

In this study, limited by its retrospective design, the prognostic value of NLR at baseline was confirmed in advanced STS patients, irrespective of treatment. Changes in NLR during the first 50 days of treatment with pazopanib were not associated with patient outcome and can therefore not be used as a marker for response.