EORTC

The future of cancer therapy

EORTC experience with advanced/metastatic epithelioid sarcoma patients treated in prospective trials: clinical profile and response to systemic therapy

Abstract 2110

Touati N[1], Schöffski P[2], Litière S[1], Judson I[3], Sleijfer S[4], van der Graaf WT[5], Italiano A[6], Isambert N[7], Gil T[8], Blay JY[9], Stark D[10], Brodowicz T[11], Marréaud S[1], Gronchi A[12]

[1]European Organization for Research and Treatment of Cancer, Brussels, Belgium; [2]University Hospitals Leuven, KU Leuven, Belgium; [3]Royal Marsden Hospital, London, UK; [4]Erasmus MC - Cancer Institute, Rotterdam, the Netherlands; [5]The Institute of Cancer Research and the Royal Marsden NHS Foundation Trust, London, UK; [6]Institut Bergonié, Bordeaux, France; [7]Centre Georges-François Leclerc, Dijon, France; [8]Jules Bordet Institute, Université Libre de Bruxelles, Brussels, Belgium; [9]Centre Léon Bérard, Lyon, France; [10]Leeds Teaching Hospitals NHS Trust - St. James University Hospital, Leeds, UK; [11]Medical University Vienna - General Hospital, Vienna, Austria; [12]Fondazione IRCCS Istituto Nazionale dei Tumori, Milano, Italy

Table 1: Patient and tumor characteristics

Characteristics		Total (N=27) N(%)
Treatment line	**1st line treatment**	18 (66.7)
	Doxorubicin	*5*
	Doxorubicin + ifosfamide	*8*
	Pazopanib	*2*
	Trabectedin	*3*
	2nd + line treatment *(pazopanib)*	9 (33.3)
Gender	Male	17 (63.0)
	Female	10 (37.0)
Site of primary tumor	Lower extremity	8 (29.6)
	Upper extremity	5 (18.5)
	Retro-intra abdominal	4 (14.8)
	Thoracic	2 (7.4)
	Visceral Gynecological	2 (7.4)
	Head and neck	1 (3.7)
	Trunk	1 (3.7)
	Visceral Breast	1 (3.7)
	Other	3 (11.1)
Performance status	0	16 (59.3)
	1	11 (40.7)
Extension of disease at entry*	Lung	17 (63.0)
	Primary	15 (55.6)
	Lymph nodes	9 (33.3)
	Bone	8 (29.6)
	Soft tissue	7 (25.9)
	Skin invasive	4 (14.8)
	Pleural effusion	4 (14.8)
	Liver	2 (7.4)
	Other site	4 (14.8)

Percentages are not cumulative

BACKGROUND

Epithelioid sarcoma (ES) is a soft-tissue sarcoma (STS) associated with a high local recurrence rate after primary resection and high incidence of distant metastasis. Little is known about clinical course and response to systemic treatments. This retrospective analysis aims to provide a reference for future ES-specific studies.

PATIENT AND METHODS

Data from patients with locally advanced/metastatic ES entered in prospective multi-sarcoma phase II/III trials were pooled: EORTC trial 62012 (doxorubicin vs. doxorubicin/ifosfamide), 62043 (pazopanib), 62072 (pazopanib vs. placebo) and 62091 (doxorubicin vs. trabectedin). Patients had either a local or centrally confirmed diagnosis of ES, inoperable/metastatic disease at study entry and were eligible for the respective trial. Response was assessed using RECIST 1.1. Progression-free survival (PFS) and overall survival (OS) were calculated from date of study entry.

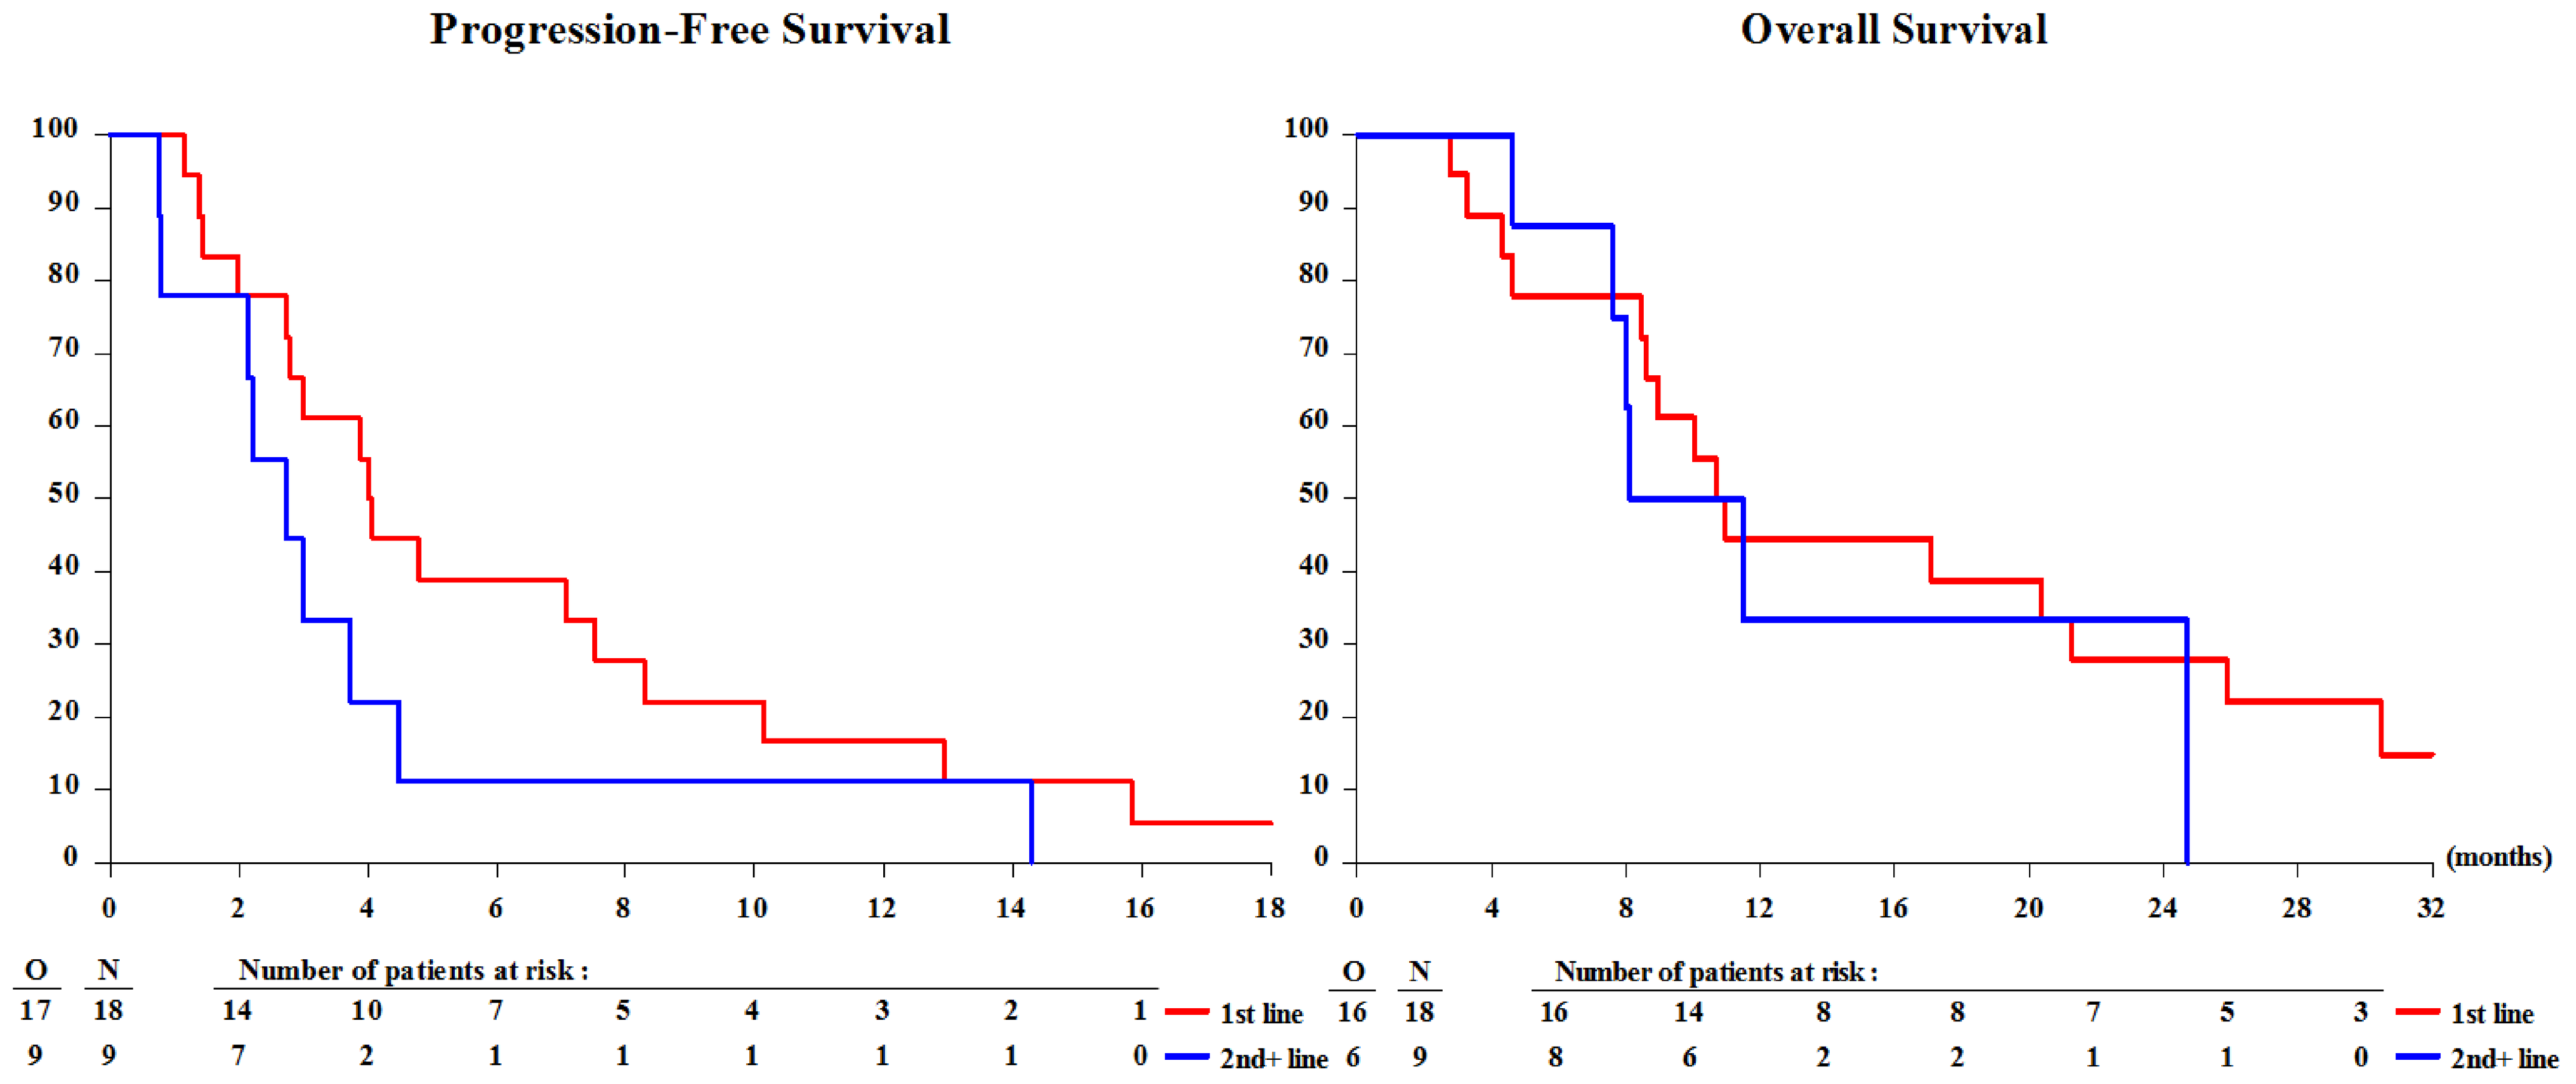

Progression-Free Survival
Overall Survival
100
90
80
70
60
50
40
30
20
10
0
0 2 4 6 8 10 12 14 16 18
0 4 8 12 16 20 24 28 32
(months)
O N Number of patients at risk :
17 18 14 10 7 5 4 3 2 1 1st line
9 9 7 2 1 1 1 1 1 0 2nd+ line
16 18 16 14 8 8 7 5 3 1st line
6 9 8 6 2 2 1 1 0 2nd+ line

Figure 1: Progression-free survival (left) and overall survival (right) by line of treatment; 1st line of treatment in red and 2nd line or later in blue

Table 2: Progression-free survival, overall survival and objective response rate according to the line of treatment and overall

	Patients	Observed PD/deaths*	Median PFS (95% CI; Months)	Observed deaths	Median OS (95% CI; Months)	Objective Response rate
1st line treatment	18	17	4.04 (2.73, 7.52)	16	10.83 (8.44, 21.29)	**4/18 = 22.2%**
2nd + line treatment	9	9	2.73 (0.76, 4.47)	6	9.79 (4.63, 24.71)	**1/9 = 11.1%**
All patients	27	26	3.75 (2.20, 4.80)	22	10.83 (8.11, 21.29)	**5/27 = 18.5%**

**: whichever came first*

RESULTS

Among 1099 patients with advanced STS in the pooled studies, 27 ES patients (2.5%) were eligible for the current retrospective analysis. Patient and tumor characteristics are illustrated in Table 1. The tumor grade was assessed for 21 patients and characterized as high for 10 patients, intermediate for 10 and low for 1. Best response to the different drugs used for 1st line patients was 4 partial responses (PR, 22.2%), 10 stable disease (SD, 55.6%) and 4 progressive disease (PD, 22.2%). In subsequent lines, pazopanib achieved 1 PR (11.1%), 4 SD (44.4%) and 4 PD (44.4%).

Patient's prognosis and response rate are detailed in Table 2. Median follow-up was 45.3 months and all patients but one progressed. Five patients were still alive at time of the according trial analysis and 13 patients died because of progressive disease.

CONCLUSION

With all limitations of a retrospective analysis of such small dataset, objective response and survival outcomes of this locally advanced/metastatic ES population are relatively poor. The clinical testing of novel agents remains a high priority in this sarcoma subtype.

Funding: *this study was done by EORTC Headquarters and funded by Epizyme*